7

Presented at the

Dermatology Nurses' Association 40th Annual Convention

Las Vegas, NV, USA
February 23–25, 2022

Exploring the Natural and Treatment History of Vitiligo: Findings From the Global VALIANT Study

Kristen Bibeau, PhD, MSPH,[1] Davinder Parsad, MD,[2] John E. Harris, MD, PhD,[3] Yan Valle, MSc, MBA,[4] Mukta Tulpule, MBBS,[5] Gaone Tlhong Matewa, BBA,[6] Christine LaFiura, BA,[7] Anouk Lindley, MBA,[1] Khaled Ezzedine, MD, PhD[8]

[1]Incyte Corporation, Wilmington, DE, USA; [2]Post Graduate Institute of Medical Education and Research, Chandigarh, India; [3]University of Massachusetts Medical School, Worcester, MA, USA; [4]Vitiligo Research Foundation, New York, NY, USA; [5]Shweta Association, Pune, India; [6]Beyond Vitiligo, Johannesburg, South Africa; [7]Envision Health Partners LLC, Riverside, CT, USA; [8]Henri Mondor University Hospital and Université Paris-Est Créteil Val de Marne, Paris, France

Introduction

- Vitiligo is a chronic autoimmune disease characterized by the destruction of melanocytes, resulting in pale or white patches of skin[1]
- It is estimated that vitiligo affects 0.5%–2% of the global population, with rates varying geographically[2]
- Vitiligo affects adults and children of both sexes equally[3]; disease onset commonly occurs by 30 years of age, although initial manifestations later in life are also common[4,5]
- Extensive body surface area (BSA) involvement, darker skin types, and the presence of visible lesions have been associated with greater patient burden[6]
- There is a need to understand the experiences among patients with vitiligo from different countries and with different disease characteristics

Objective

- The population-based Vitiligo and Life Impact Among International Communities (VALIANT) study sought to understand the natural history of vitiligo among patients around the world, as well as the patient journey with vitiligo

Methods

Study Design and Patients

- This cross-sectional online survey recruited adult patients (aged ≥18 years) diagnosed with vitiligo by a healthcare professional
- Patients were recruited using a general population sampling approach from a network of consumers in the United States, Europe (France, Germany, Italy, Spain, United Kingdom), Canada, Australia, Brazil, Asia (China, India, Japan, Philippines, Thailand), and Africa/Middle East (Egypt, Saudi Arabia, South Africa)
- Patients completed a self-administered online screener designed to capture high-level demographics, confirm diagnosed vitiligo, and provide consent before continuing to the 25-minute survey
- Clinical characteristics were solicited to understand the time since diagnosis of vitiligo, the diagnostic process, and the rate at which vitiligo is spreading
 - Family history of vitiligo, self-reported factors influencing vitiligo, and the type of management the patient was currently receiving were also probed
- Treatment history was examined to reflect on the use of various treatments and management strategies
- The extent of vitiligo was assessed using the validated Self Assessment Vitiligo Extent Score (SA-VES) tool,[7] which uses an array of validated images for the patient to self-select, indicating how many vitiligo lesions are on each location of the body, and estimates the affected BSA

Statistical Analyses

- Data were analyzed using descriptive statistics, with mean (SD) and median (range) for continuous variables, and percentages for discrete variables
- Statistical comparisons were made between subgroups (eg, fair vs dark skin), with significance conferred at the level of P<0.05; no corrections were made for multiple testing

Results

Patient Demographics

- Of 881,522 participants invited to the survey, 3919 (0.4%) completed the survey, and 3541 (0.4%) were included in the analysis
- Among the 3541 included patients, median age was 38 (18–95) years (**Table 1**)
 - More than half of the patients (54.6%) were male
 - More patients reported Fitzpatrick skin phototypes I–III (fairer skin types, 59.2%) compared with phototypes IV–VI (darker skin types, 40.8%)

Disease Characteristics

- Median (range) BSA affected by vitiligo was 4.22% (0.02%–73.88%), as measured by the SA-VES (**Table 2**)
- Mean (SD) time between first noticing lesions and receiving a formal diagnosis was 1.4 (4.1) years
- Most patients reported slow progression (36.0%) or rapid progression (31.9%) of vitiligo, where only 8.4% reported no progression since the first appearance of lesions
- Patients with high BSA involvement (>5%) were significantly (P<0.05) younger at first appearance of lesions (mean [SD], 27.3 [12.9] years) than those with medium (1%–5%, 29.3 [12.6] years) or low (<1%, 31.8 [14.4] years) BSA involvement

Table 1. Patient Demographics

Characteristic	All Participants (N=3541)
Age, median (range), y	38 (18–95)
Age range, n (%), y	
18–34	1280 (36.1)
35–54	1543 (43.6)
≥55	718 (20.3)
Male, n (%)	1933 (54.6)
Geographic region, n (%)	
United States	608 (17.2)
Europe	1151 (32.5)
Canada	200 (5.6)
Australia	75 (2.1)
Brazil	301 (8.5)
Asia	1005 (28.4)
Africa/Middle East	201 (5.7)
Race,*† n (%)	
White	1555 (51.1)
Asian	929 (30.5)
Black	283 (9.3)
Other	354 (11.6)
Fitzpatrick skin phototype,‡ n (%)	
I	317 (9.0)
II	1061 (30.0)
III	718 (20.3)
IV	817 (23.1)
V	525 (14.8)
VI	103 (2.9)

* Multiple answers were accepted.
† Race was not solicited in France (n=250) or Germany (n=250).
‡ Fitzpatrick skin phototypes are defined as follows: type I, pale white skin; type II, white skin; type III, light brown skin; type IV, moderate brown skin; type V, dark brown skin; type VI, deeply pigmented dark brown to black skin.

Table 2. Disease Characteristics

Characteristic	All Participants (N=3541)
Age at diagnosis, median (range), y	30 (1–95)
Disease duration, mean (SD), y	11.7 (12.6)
Affected BSA, median (range), %	4.22 (0.02–73.88)
Affected BSA range, n (%)	
<1% (low)*	984 (27.8)
1%–5% (medium)	955 (27.0)
>5% (high)	1602 (45.2)
Disease progression, n (%)	
No progression	299 (8.4)
Slow progression	1274 (36.0)
Stable, then rapid	804 (22.7)
Rapid, no stabilization	414 (11.7)
Rapid at first, then stabilized	524 (14.8)
Rapid, short bursts separated by stabilization	190 (5.4)
Other	36 (1.0)
Stress-induced vitiligo flares, n (%)	2339 (66.1)
Experience itch before or during flares, n (%)	2309 (65.2)
Family history, n (%)	2022 (57.1)

BSA, body surface area.
* 87 patients reported a BSA of zero.

- Nearly half (45.2%) of patients had high BSA involvement, with significantly higher rates in Africa/Middle East (58.7% of 201 patients) compared with other regions (**Figure 1**)
 - Patients with darker skin types (55.3% vs 38.3% for fairer skin) or facial lesions (63.2% vs 18.9% no facial lesions) had significantly greater rates of high BSA involvement

Figure 1. Patients With >5% Affected BSA

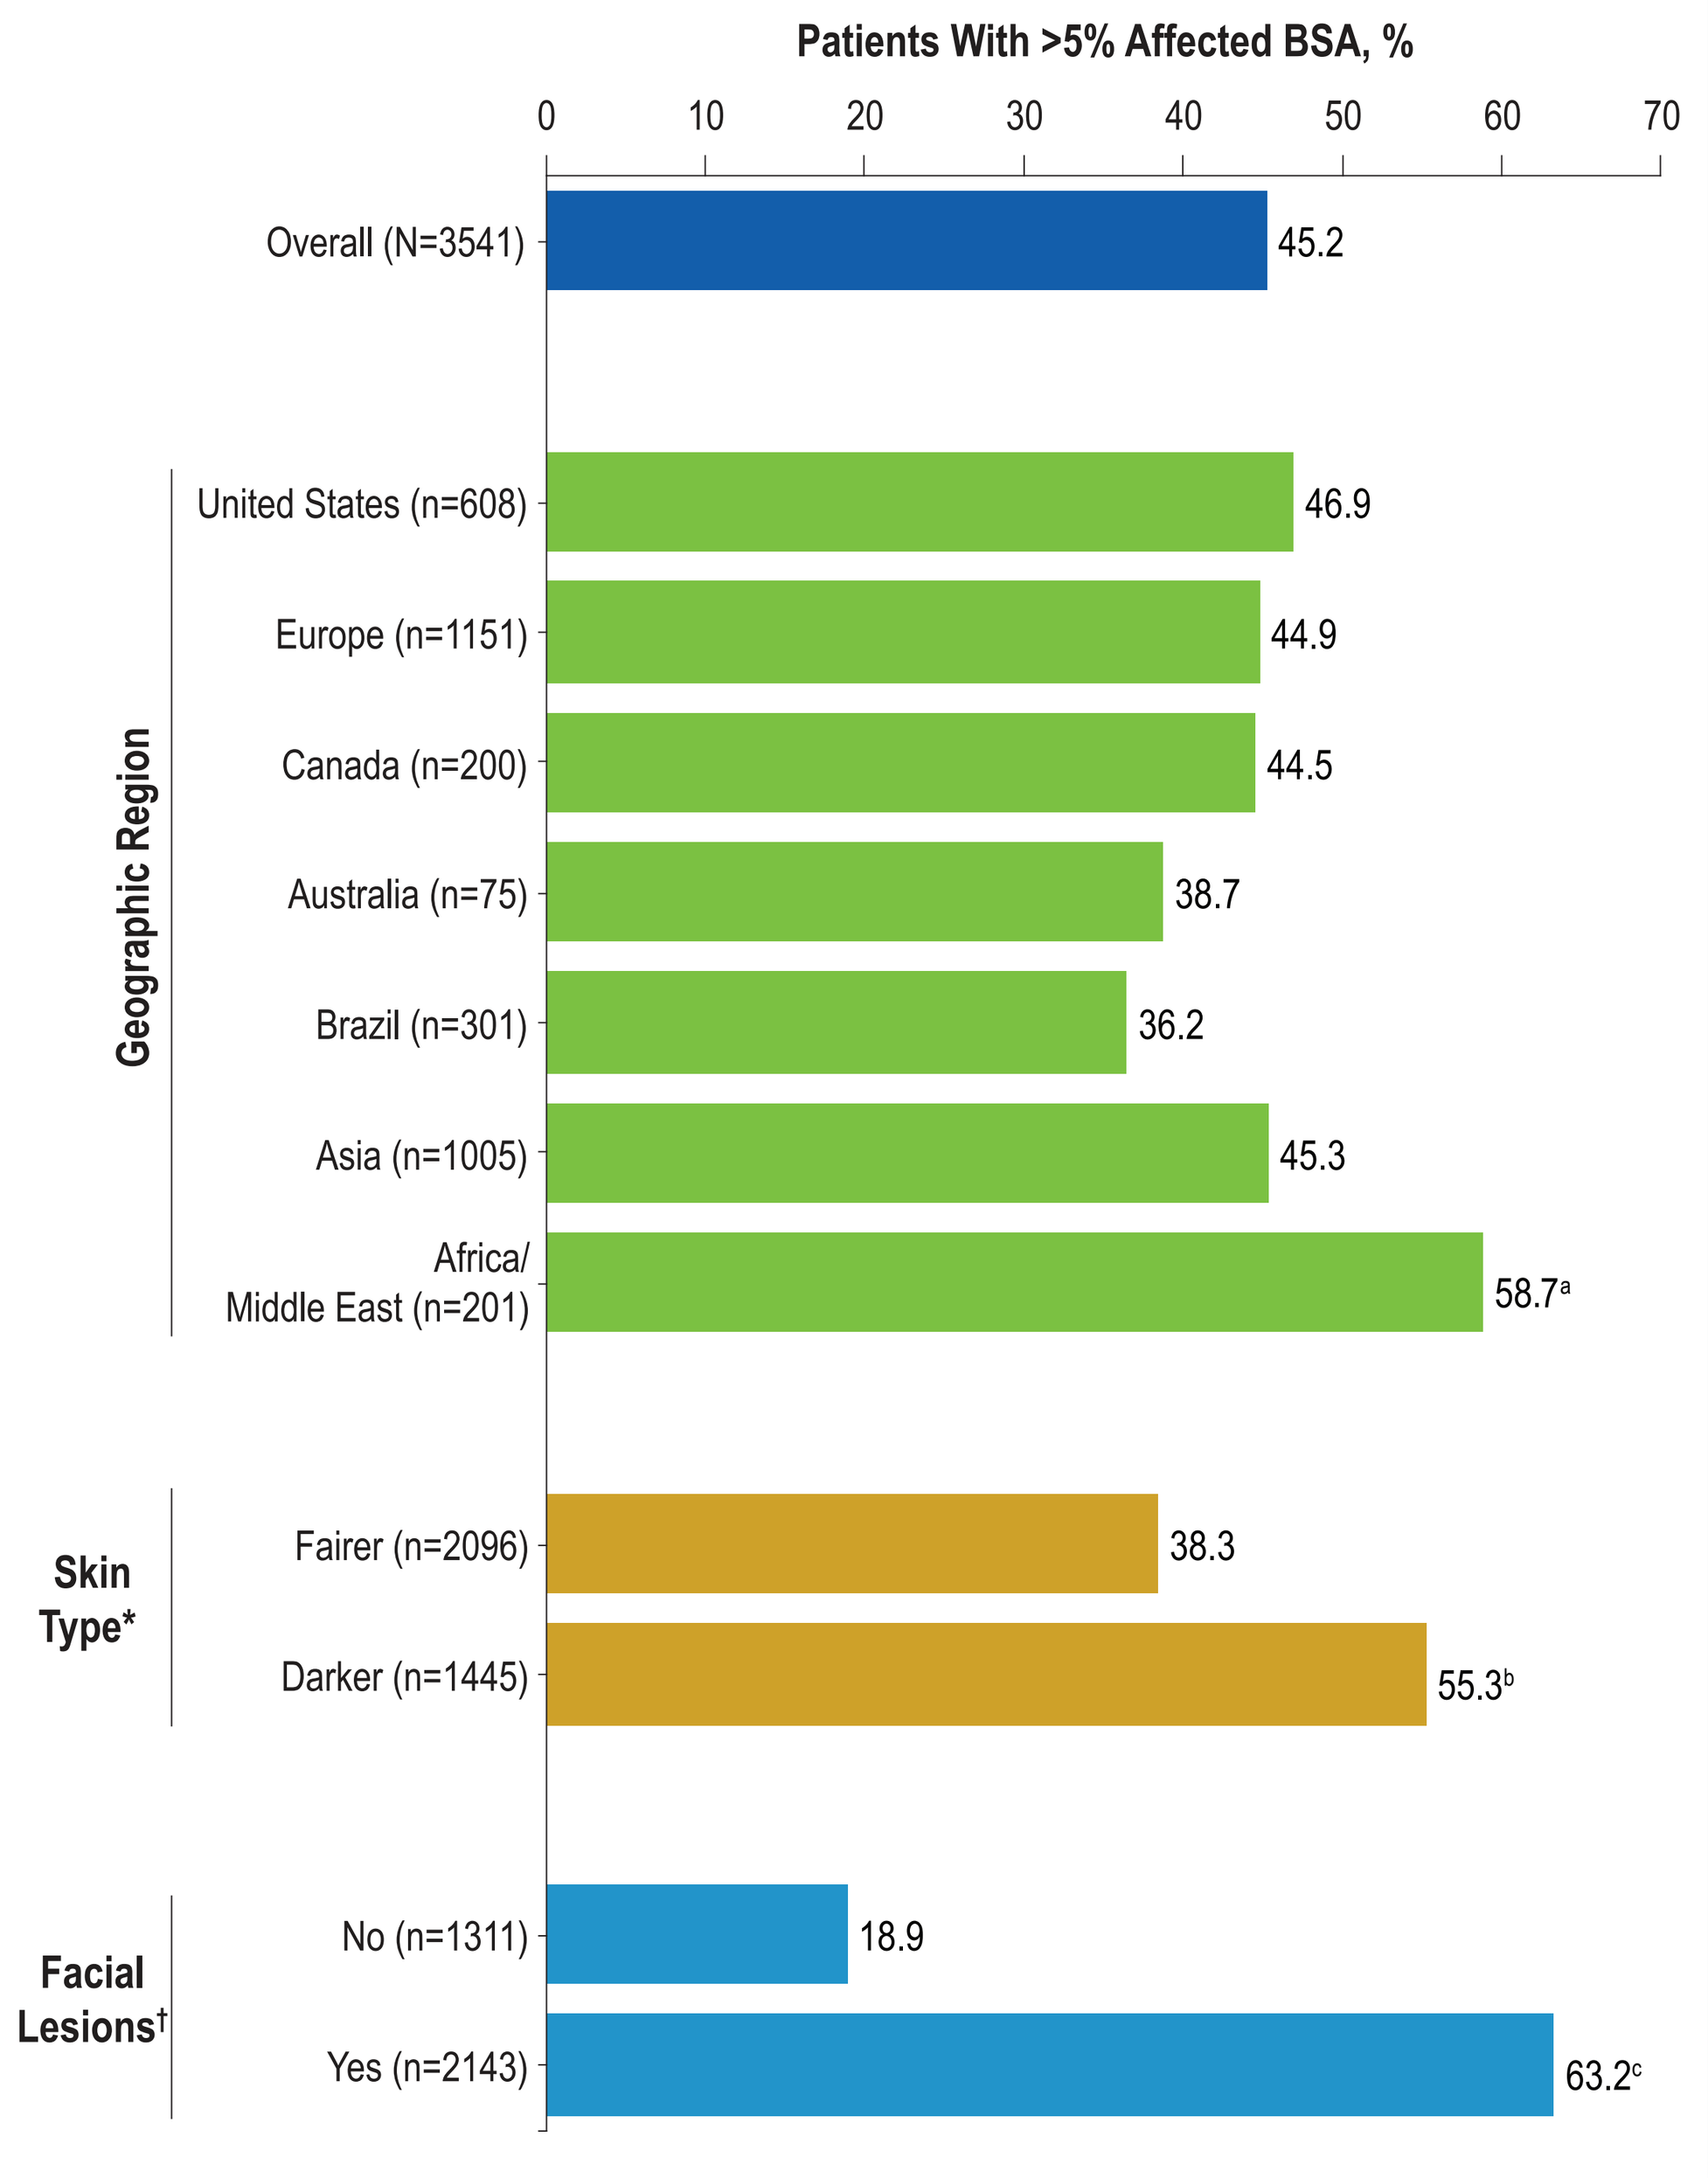

Patients With >5% Affected BSA, %
0
10
20
30
40
50
60
70
Overall (N=3541) 45.2
Geographic Region
United States (n=608) 46.9
Europe (n=1151) 44.9
Canada (n=200) 44.5
Australia (n=75) 38.7
Brazil (n=301) 36.2
Asia (n=1005) 45.3
Africa/ Middle East (n=201) 58.7[a]
Skin Type*
Fairer (n=2096) 38.3
Darker (n=1445) 55.3[b]
Facial Lesions†
No (n=1311) 18.9
Yes (n=2143) 63.2[c]

BSA, body surface area.
* Fitzpatrick skin phototypes I–III were characterized as fairer and phototypes IV–VI as darker skin types.
† Patients with 0% affected BSA (n=87) were excluded from analysis of facial lesions.
[a] P<0.05 vs other geographic regions; [b] P<0.05 vs fairer skin types; [c] P<0.05 vs no facial lesions.

- Almost three-fifths of patients (57.1%) noted a family history of vitiligo, which was most common among patients with high BSA involvement (71.2% vs 52.4% for medium and 38.8% for low), darker skin types (69.0% vs 48.9% for fairer skin), and facial lesions (64.3% vs 45.0% no facial lesions)
- Two-thirds (66.1%) of patients noted experiencing flares during periods of stress, and 65.2% noticed itching before or during a flare (**Figure 2**)
 - Significantly higher rates of flares during periods of stress were reported by patients with high BSA involvement (83.6% vs 61.5% for medium and 41.9% for low), darker skin types (76.1% vs 59.1% for fairer skin), or facial lesions (75.2% vs 51.7% for no facial lesions)
 - Similarly, significantly higher rates of experiencing itch before or during a flare were reported by patients with high BSA involvement (80.1% vs 63.4% for medium and 42.7% for low), darker skin types (73.4% vs 59.6% for fairer skin), or facial lesions (72.5% vs 53.8% no facial lesions)

Figure 2. Patient Experiences of (A) Flares During Periods of Stress and (B) Itching Before or During Vitiligo Flares

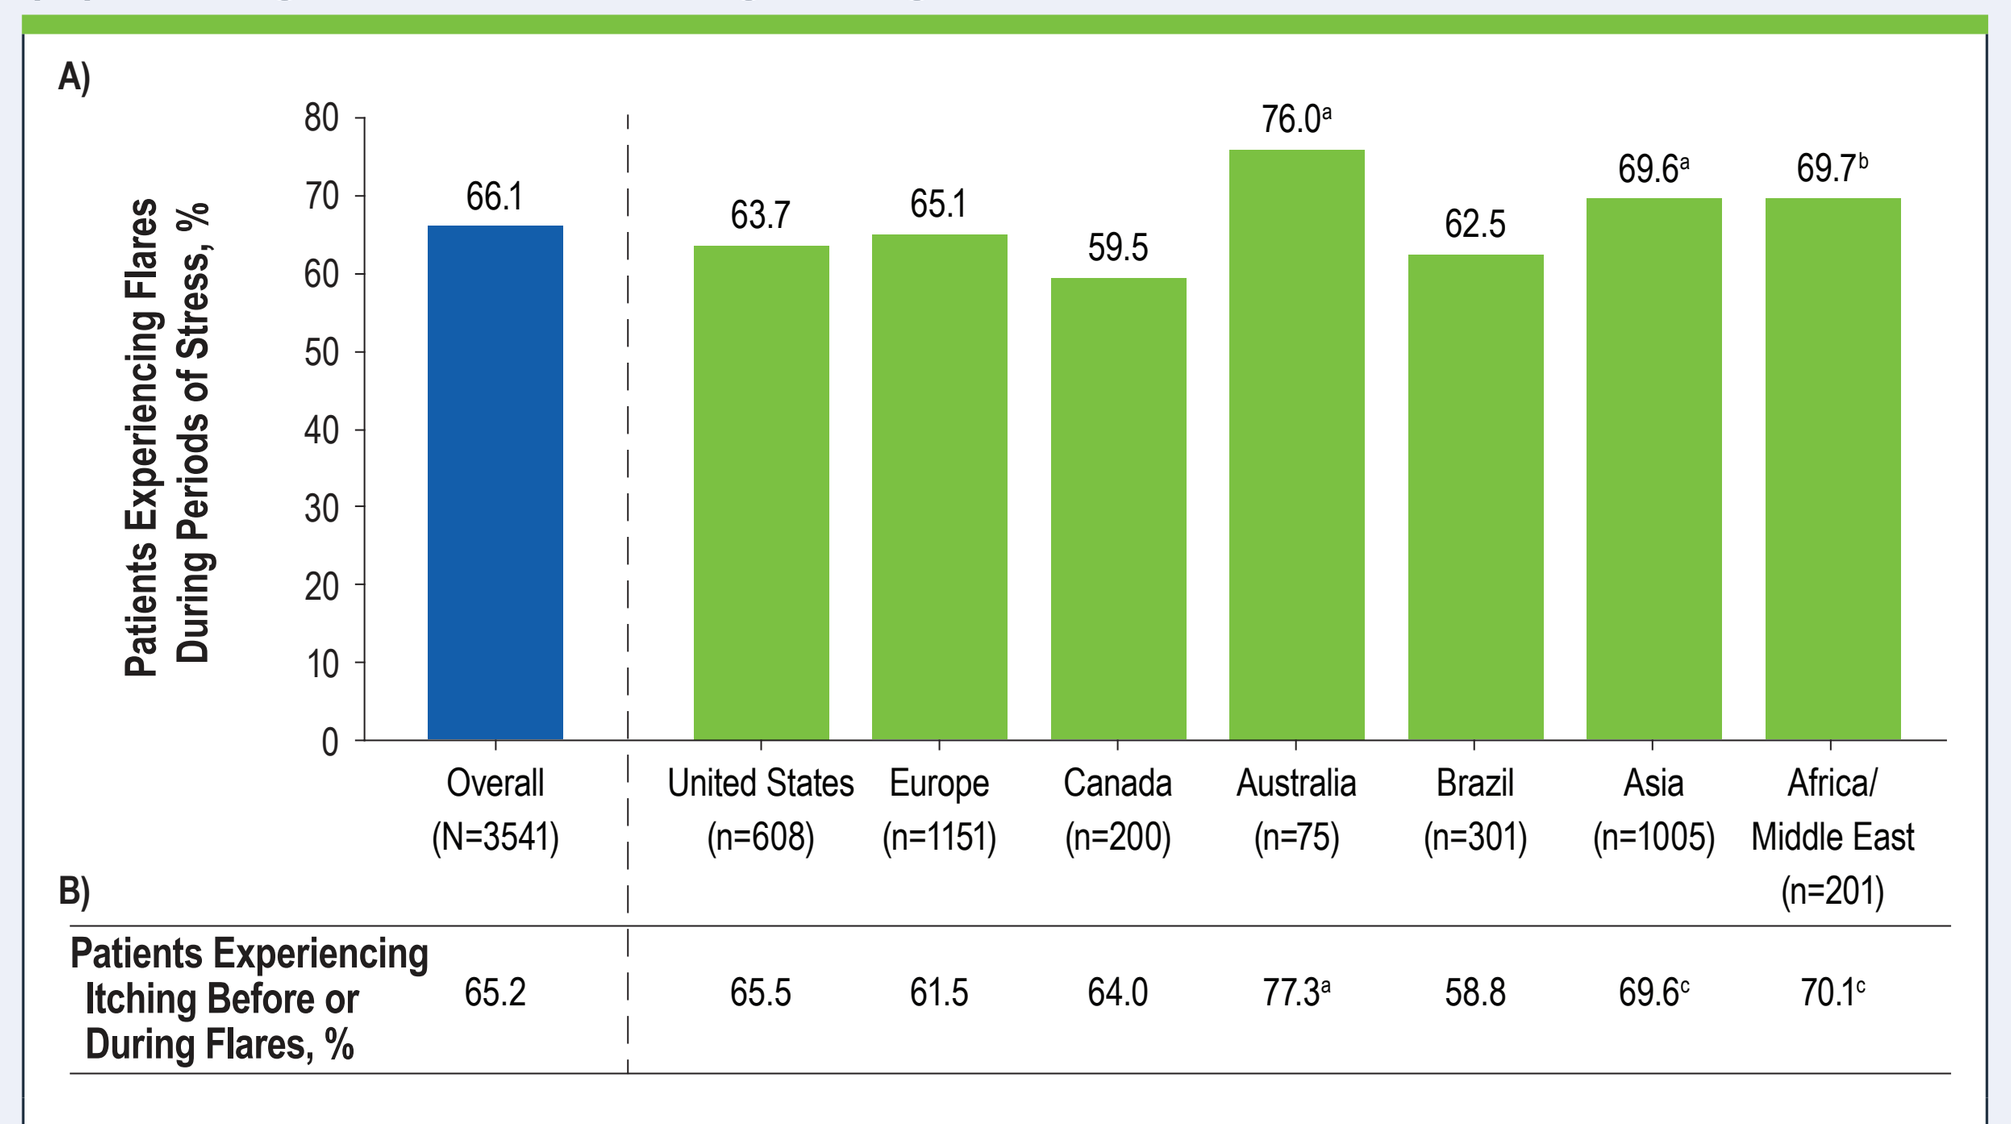

A)
Patients Experiencing Flares During Periods of Stress, %
0
10
20
30
40
50
60
70
80
66.1
63.7
65.1
59.5
76.0[a]
62.5
69.6[a]
69.7[b]
Overall (N=3541)
United States (n=608)
Europe (n=1151)
Canada (n=200)
Australia (n=75)
Brazil (n=301)
Asia (n=1005)
Africa/ Middle East (n=201)
B)
Patients Experiencing Itching Before or During Flares, %
65.2
65.5
61.5
64.0
77.3[a]
58.8
69.6[c]
70.1[c]

[a] P<0.05 vs United States, Europe, Canada, and Brazil; [b] P<0.05 vs Canada; [c] P<0.05 vs Europe and Brazil.

Treatment History and Satisfaction

- Patients had used a mean (SD) of 5.9 (4.9) treatments to manage their vitiligo (**Figure 3**)
- Use of a significantly (P<0.05) greater number of treatments was reported by patients with higher BSA involvement, darker skin types, or facial lesions

Figure 3. Treatments or Management Strategies Ever Used

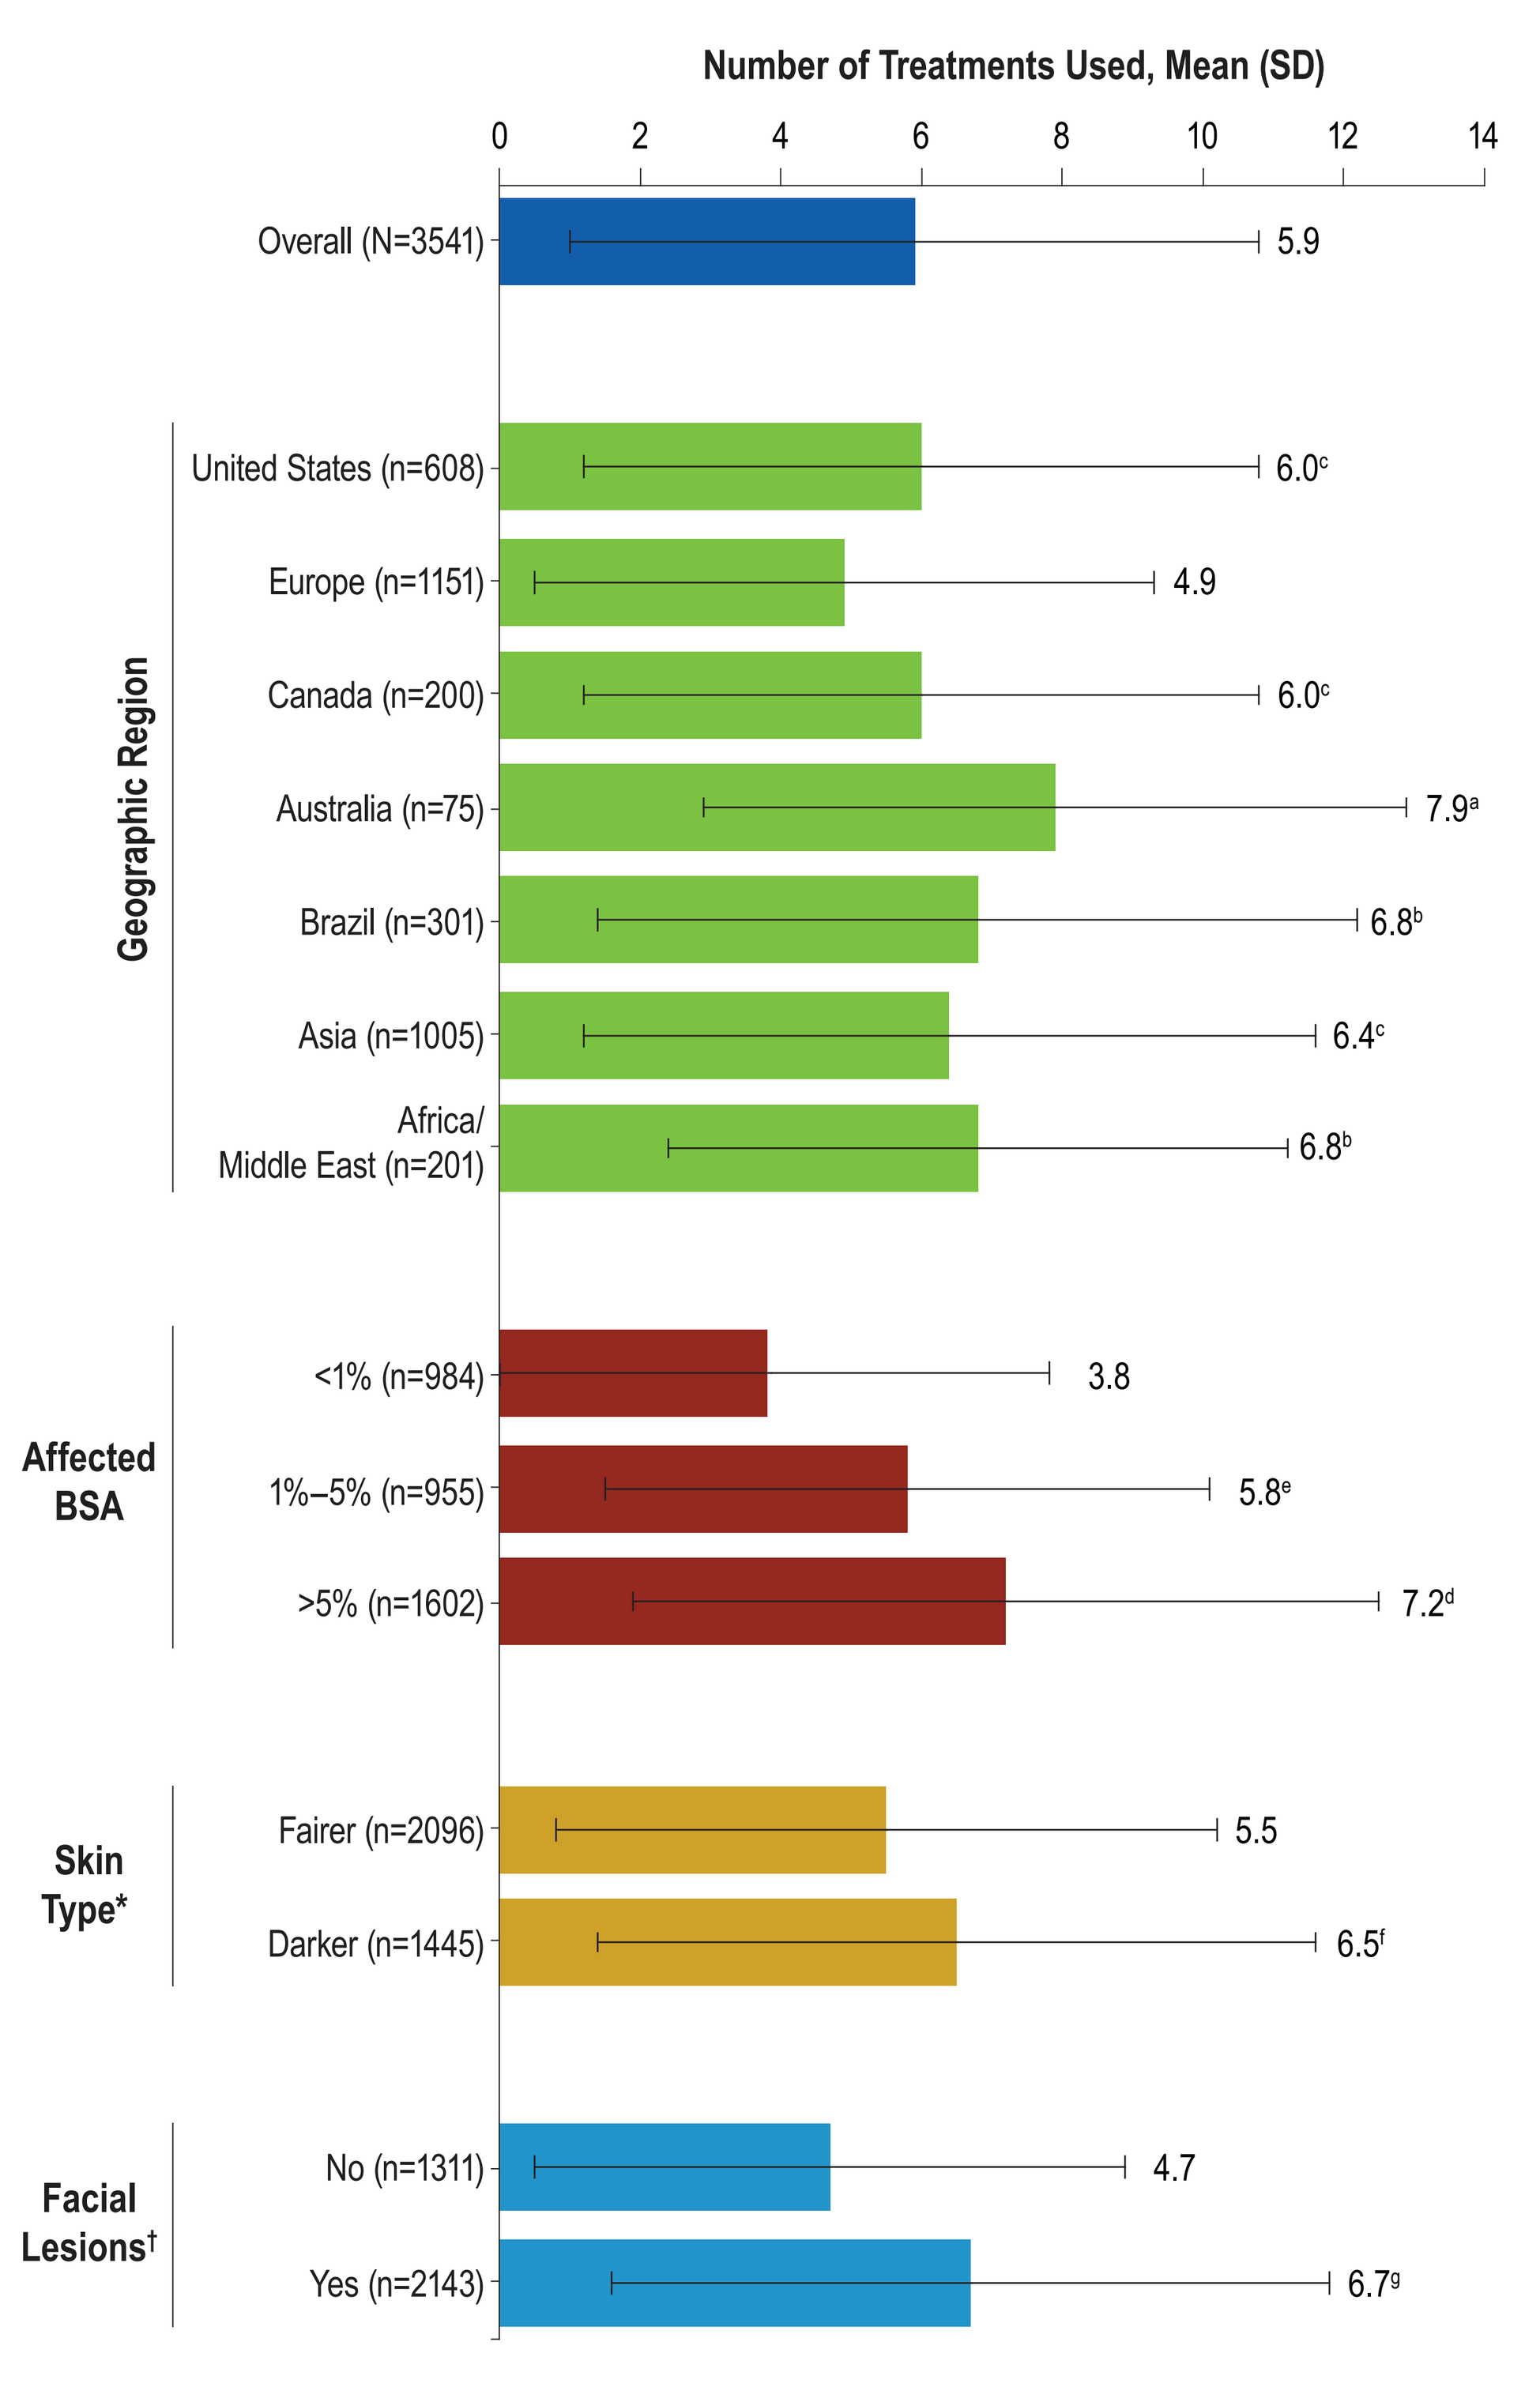

Number of Treatments Used, Mean (SD)
0
2
4
6
8
10
12
14
Overall (N=3541) 5.9
Geographic Region
United States (n=608) 6.0[c]
Europe (n=1151) 4.9
Canada (n=200) 6.0[c]
Australia (n=75) 7.9[a]
Brazil (n=301) 6.8[b]
Asia (n=1005) 6.4[c]
Africa/ Middle East (n=201) 6.8[b]
Affected BSA
<1% (n=984) 3.8
1%–5% (n=955) 5.8[e]
>5% (n=1602) 7.2[d]
Skin Type*
Fairer (n=2096) 5.5
Darker (n=1445) 6.5[f]
Facial Lesions†
No (n=1311) 4.7
Yes (n=2143) 6.7[g]

BSA, body surface are.
* Fitzpatrick skin phototypes I–III were characterized as fairer and phototypes IV–VI as darker skin types.
† Patients with 0% affected BSA (n=87) were excluded from analysis of facial lesions.
[a] P<0.05 vs United States, Europe, Canada, and Asia; [b] P<0.05 vs United States and Europe; [c] P<0.05 vs Europe; [d] P<0.05 vs 1%–5% and <1% affected BSA; [e] P<0.05 vs <1% affected BSA; [f] P<0.05 vs fairer skin types; [g] P<0.05 vs no facial lesions.

- Among prescription treatment options, the use of topical creams or ointments (62.2%) was more common than oral treatments (41.9%; **Figure 4**)
 - 8.0% of patients were treatment-naive

Figure 4. Current and/or Previous Treatments or Management Strategies*

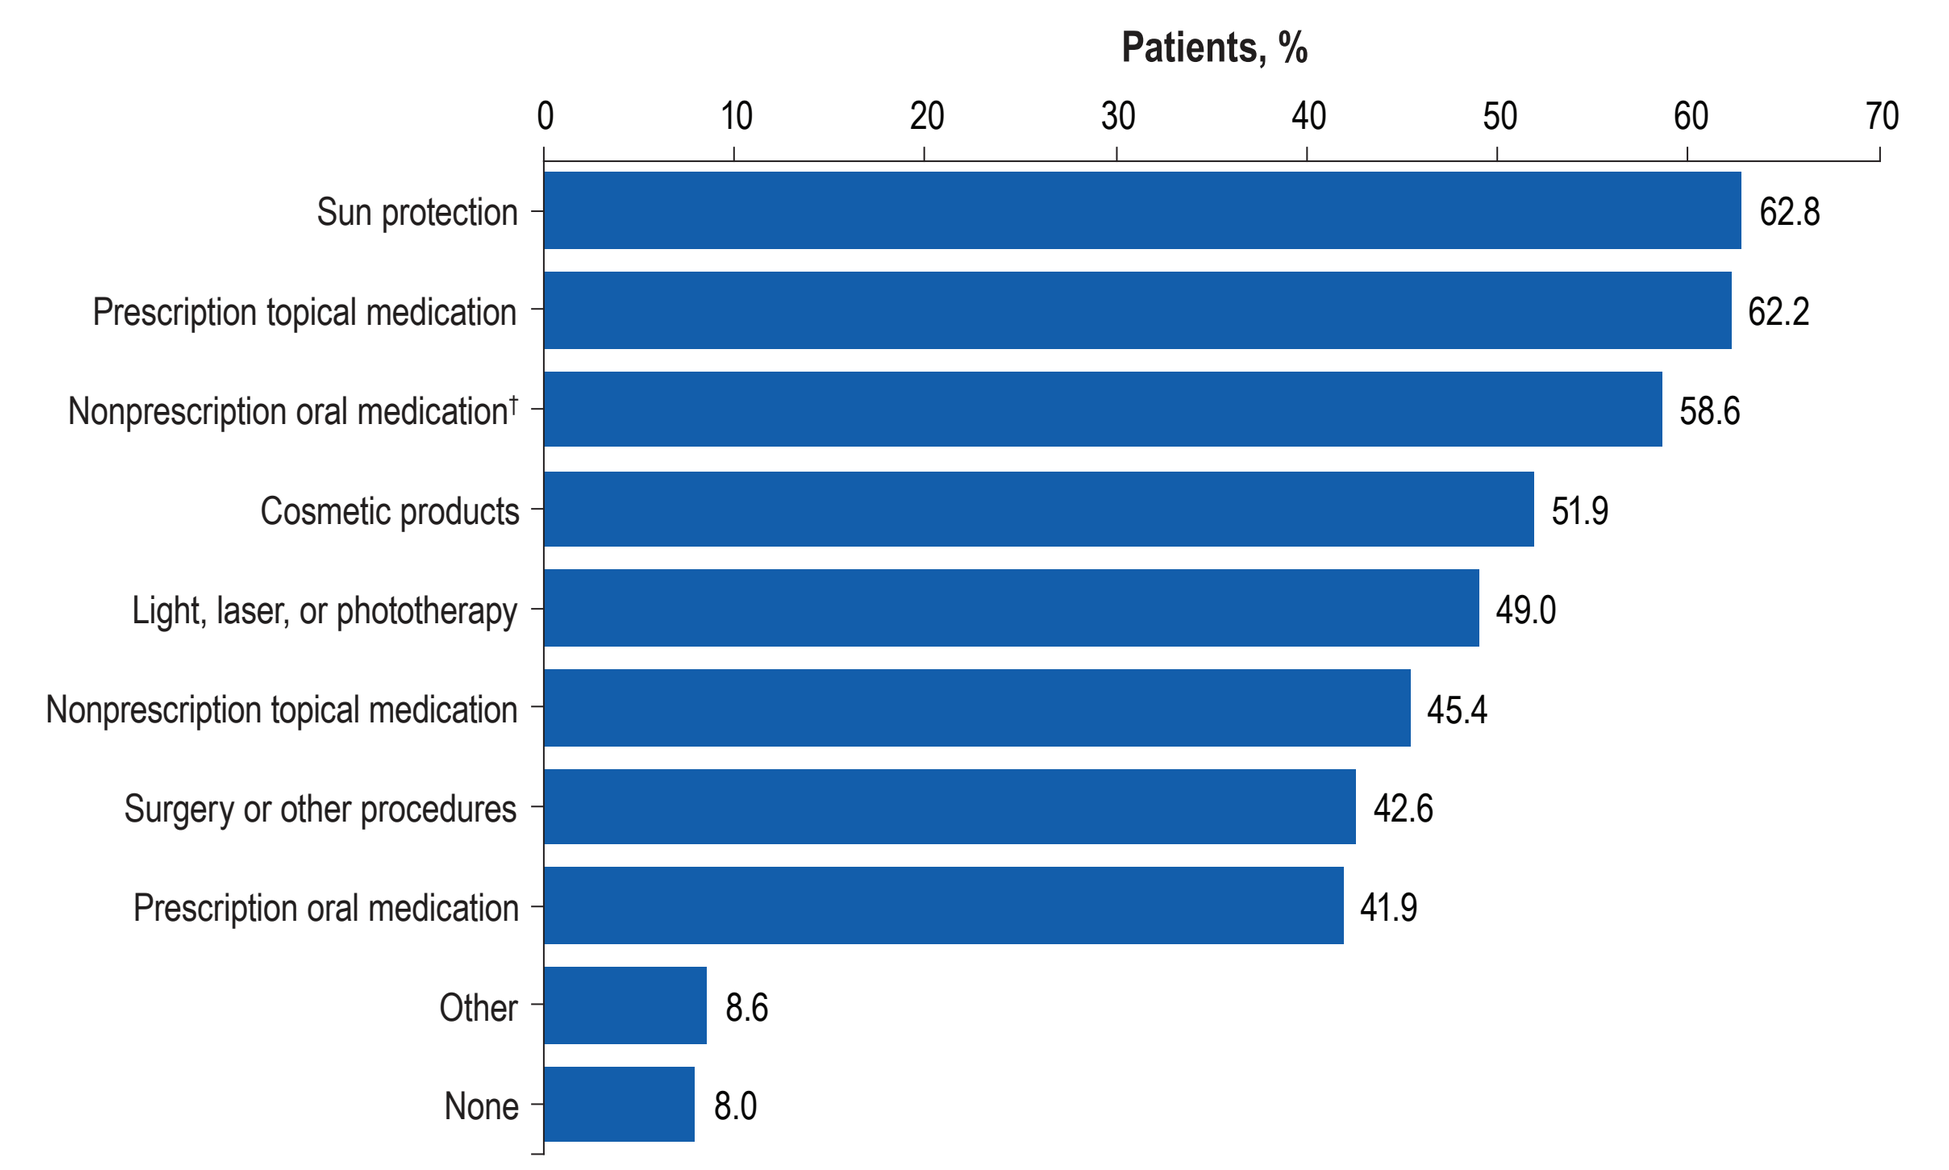

Patients, %
0
10
20
30
40
50
60
70
Sun protection 62.8
Prescription topical medication 62.2
Nonprescription oral medication† 58.6
Cosmetic products 51.9
Light, laser, or phototherapy 49.0
Nonprescription topical medication 45.4
Surgery or other procedures 42.6
Prescription oral medication 41.9
Other 8.6
None 8.0

* Patients could select multiple treatments or management strategies.
† Medications include vitamins A and D and other oral nonprescription antioxidants, vitamins or supplements.

- Globally, 14.1% of patients were not actively using treatment, with significantly higher rates in Brazil (21.9% of 301 patients) compared with other regions (**Figure 5A**)
- Most patients (68.7%) were hopeful that a new treatment will become available, with the greatest proportions in Asia (75.1% of 1005 patients) and Africa/Middle East (71.6% of 201 patients; **Figure 5B**)

Figure 5. Patients (A) Not Actively Using Treatment and (B) Hopeful That a New Treatment Will Become Available

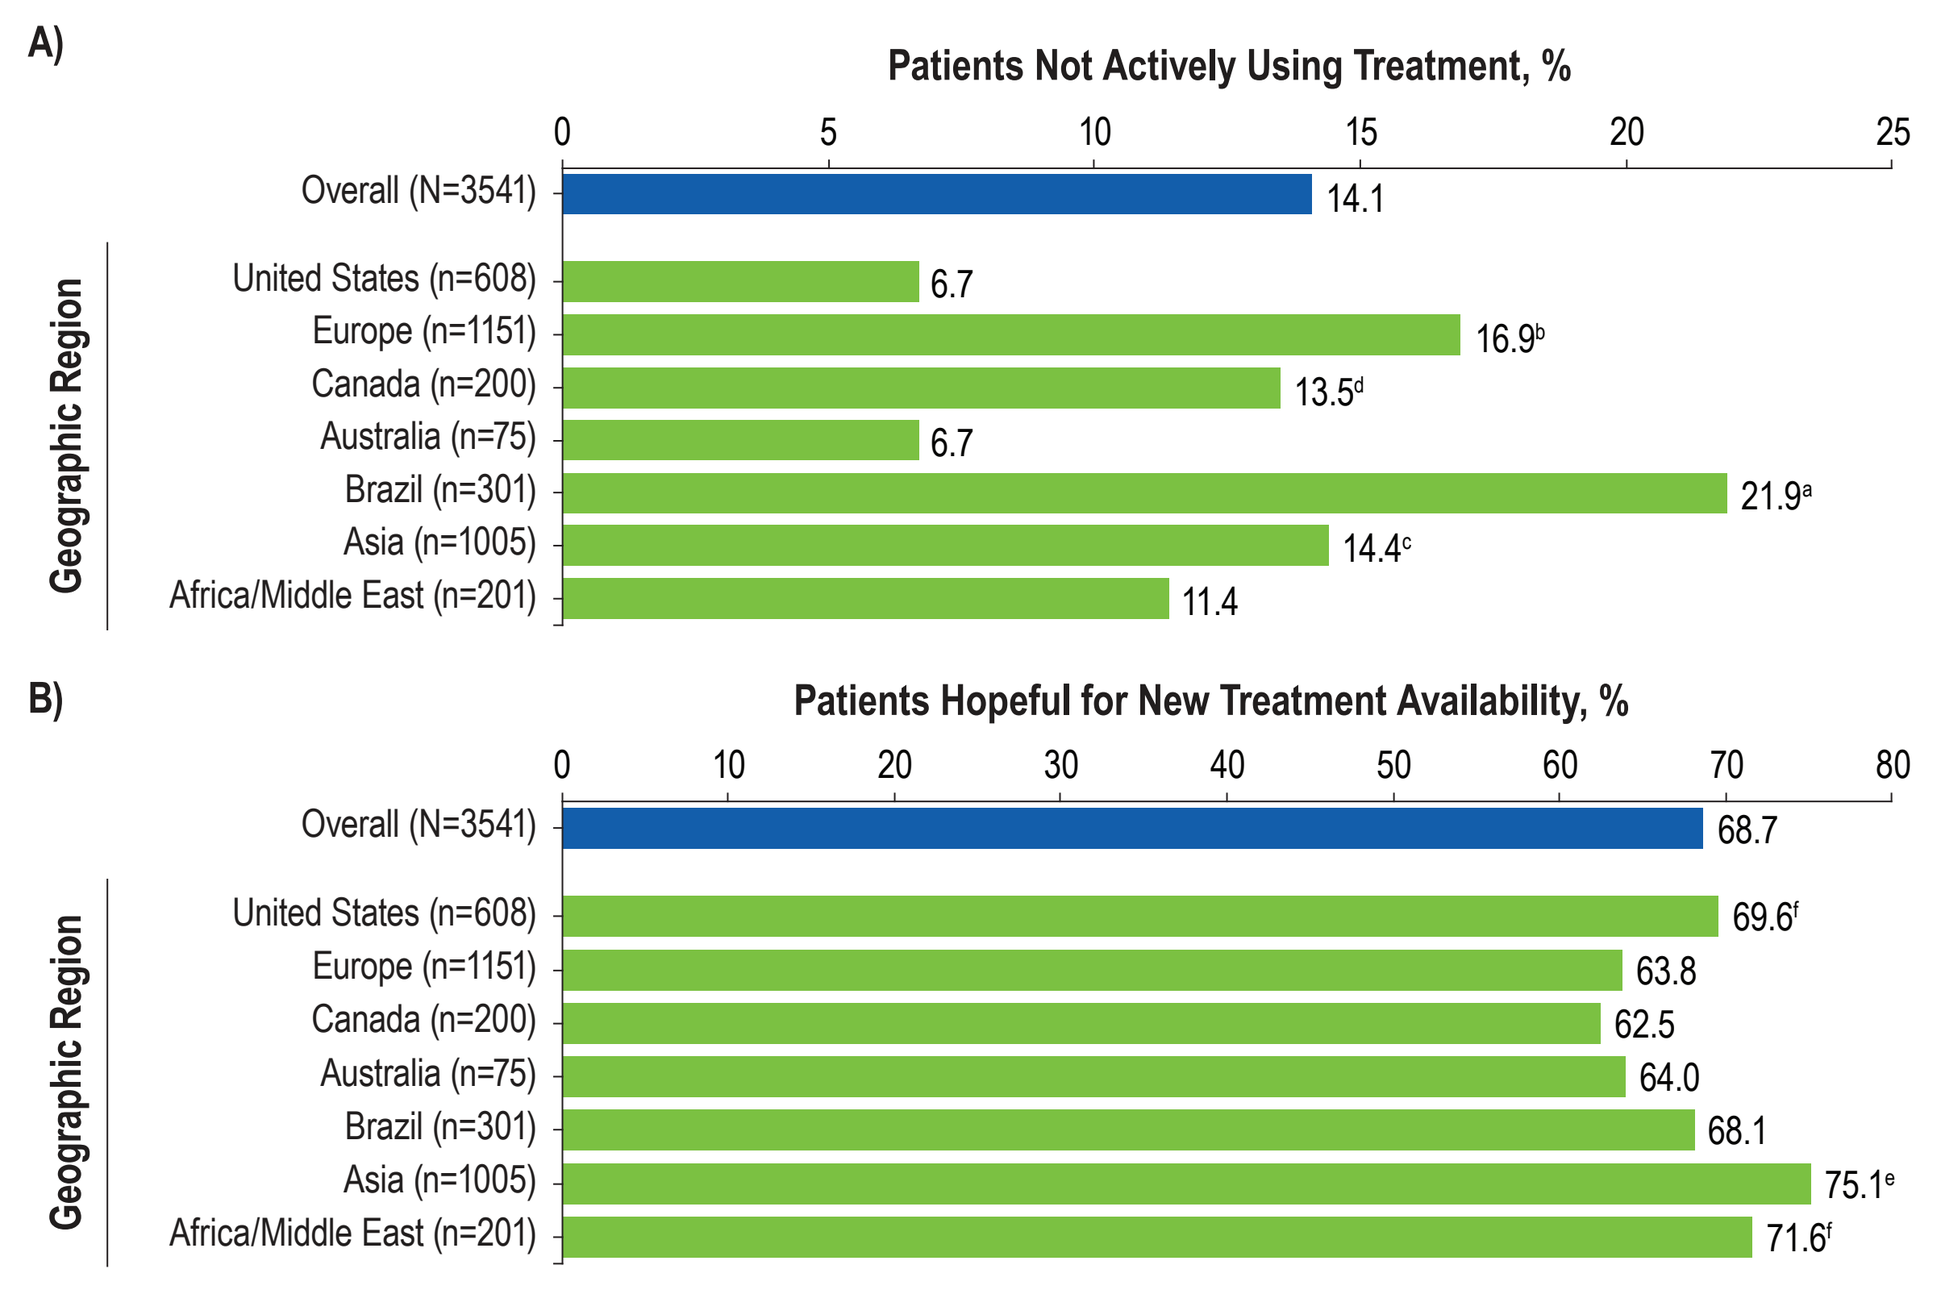

A)
Patients Not Actively Using Treatment, %
0
5
10
15
20
25
Overall (N=3541) 14.1
Geographic Region
United States (n=608) 6.7
Europe (n=1151) 16.9[b]
Canada (n=200) 13.5[d]
Australia (n=75) 6.7
Brazil (n=301) 21.9[a]
Asia (n=1005) 14.4[c]
Africa/Middle East (n=201) 11.4
B)
Patients Hopeful for New Treatment Availability, %
0
10
20
30
40
50
60
70
80
Overall (N=3541) 68.7
Geographic Region
United States (n=608) 69.6[f]
Europe (n=1151) 63.8
Canada (n=200) 62.5
Australia (n=75) 64.0
Brazil (n=301) 68.1
Asia (n=1005) 75.1[e]
Africa/Middle East (n=201) 71.6[f]

[a] P<0.05 vs United States, Canada, Australia, Asia, and Africa/Middle East; [b] P<0.05 vs United States, Australia, and Africa/Middle East; [c] P<0.05 vs United States and Australia; [d] P<0.05 vs United States; [e] P<0.05 vs United States, Europe, Canada, and Brazil; [f] P<0.05 vs Europe.

Limitations

- Limitations are those associated with an online survey, including restriction of participants to those with internet access
 - Efforts were made to conduct in-person interviews in populations with limited internet access if needed to reach desired sample size
- Other limitations include potential errors in measurement that are inherent in patient-reported outcomes studies

Conclusions

- **On average, it took patients 1.4 years to receive a formal diagnosis of vitiligo after appearance of their first lesions**
- **Patients with higher BSA involvement (>5%) often had earlier disease onset, a family history of vitiligo, and used a greater number of treatments than other patients**
 - **High BSA involvement was more prominent among patients from Africa/Middle East, those with darker skin tones (Fitzpatrick skin phototypes IV–VI), or those with facial lesions**
- **These findings provide a new perspective on the diagnosis and treatment journey for patients with vitiligo globally**

Disclosures

KB and AL are employees and shareholders of Incyte Corporation. DP has served as an expert or primary investigator for Incyte Corporation, Pfizer, and Sun Pharmaceuticals. JEH has served as a consultant for AbbVie, Aclaris Therapeutics, BiologicsMD, EMD Serono, Genzyme/Sanofi, Janssen, Pfizer, Rheos Medicines, Sun Pharmaceuticals, TeVido BioDevices, The Expert Institute, 3rd Rock Ventures, and Villaris Therapeutics; has served as an investigator for Aclaris Therapeutics, Celgene, Dermira, EMD Serono, Genzyme/Sanofi, Incyte Corporation, LEO Pharma, Pfizer, Rheos Medicines, Stiefel/GlaxoSmithKline, Sun Pharmaceuticals, TeVido BioDevices, and Villaris Therapeutics; holds equity in Aldena Therapeutics, NIRA Biosciences, Rheos Medicines, TeVido BioDevices, and Villaris Therapeutics; is a scientific founder of Aldena Therapeutics, NIRA Biosciences, and Villaris Therapeutics; and has patents pending for IL-15 blockade for treatment of vitiligo, JAK inhibition with light therapy for vitiligo, and CXCR3 antibody depletion for treatment of vitiligo. YV is CEO of the Vitiligo Research Foundation, has served as a scientific advisor at Temprian Therapeutics, and as an invited professor at Guglielmo Marconi University. MT has no conflicts of interest to disclose. GTM is the founder of Beyond Vitiligo South Africa and cofounder of Beyond Vitiligo Botswana. CL is a co-owner of Envision Health Partners, who received funding for conducting this project from Incyte Corporation. KE is a consultant for AbbVie, Incyte Corporation, La Roche-Posay, Pfizer, Pierre Fabre, Sanofi, and Viela Bio.

Acknowledgments

This study was funded by Incyte Corporation (Wilmington, DE, USA). Writing assistance was provided by Joshua Solomon, PhD, an employee of ICON (Blue Bell, PA, USA) and was funded by Incyte Corporation.

References

1. Rodrigues M, et al. *J Am Acad Dermatol*. 2017;77(1):1-13. **2.** Kruger C and Schallreuter KU. *Int J Dermatol*. 2012;51(10):1206-1212. **3.** Ezzedine K, et al. *Lancet*. 2015;386(9988):74-84. **4.** Talsania N, et al. *Clin Exp Dermatol*. 2010;35(7):736-739. **5.** Radtke MA, et al. *Br J Dermatol*. 2009;161(1):134-139. **6.** Ezzedine K, et al. *Am J Clin Dermatol*. 2021;22(6):757-774. **7.** van Geel N, et al. *J Am Acad Dermatol*. 2017;76(3):464-471.

To download a copy of this poster, scan code.